Effects of omega-3 fatty acid on pre- and post-prandial triglyceride and metabolic parameters with standard meals in patients with hypertriglyceridemia: open, multicenter study.

1. Department of Endocrinology and Metabolism, Gangneung Asan Hospital, University of Ulsan College of Medicine, Gangneung, Republic of Korea
2. Department of Endocrinology and Metabolism, Hanyang Guri Hospital, University of Hanyang College of Medicine, Republic of Korea
3. Department of Endocrinology and Metabolism, Hallym University Sacred Heart Hospital, Hallym University School of Medicine, Republic of Korea

Won Jun Kim[1], Sangmo Hong[2], Jun Goo Kang[3], Chang Beom Lee[2]

Introduction

Usually, blood for lipid profiles are recommended to be drawn after an 8- to 12-hour fast. Because plasma triglyceride levels can increase substantially postprandially, fasting levels avoid the variability associated with meals and provide a more reliable estimate for risk assessment. However, most time of the day are likely to be exposed to the nonfasting period and postprandial lipids may play an important role in the pathogenesis of cardiovascular disease because postprandial lipids can penetrate the endothelial cell layer and contribute to the formation of foam cell(1,2). In 2007, two articles in JAMA showed nonfasting triglycerides was more important risk of cardiovascular events than fasting triglycerides(3,4). However, TG variation after meal is one of main obstacle of clinical trial. The purpose of this study is to determine effects of a 6-week period of omega-3 fatty acid supplementation on fasting and postprandial TG and metabolic parameters in response to standard test meals.

Methods

The study population included 26 patients with hypertriglyceridemia (≥ 200 mg/dl). They were educated and randomly allocated to treatment group by omega-3 fatty acid supplementation (Omacor, 2g /d) or control group, and followed-up for 6 weeks in three hospitals. The inclusion criteria of patients were as follows: age 30-75 years old, 200 mg/dL ≦ TG ≦ 499 mg/dL at screening, LDL< 200 mg/dL, no change of medication during 3 months. The exclusion criteria were as follows:, taking medication that affect lipid profiles except statin, abnormality of TFT, LFT or creatinine and alcohol consumption ≥ 30 units per week.

At day 1 and day 42 visit, fasting and postprandial-3hr data were obtained respectively, before and after standard meals. Samples were obtained two times at fasting (around 09.00 hours) and postprandial period (around 12:00 hours) at each visit.

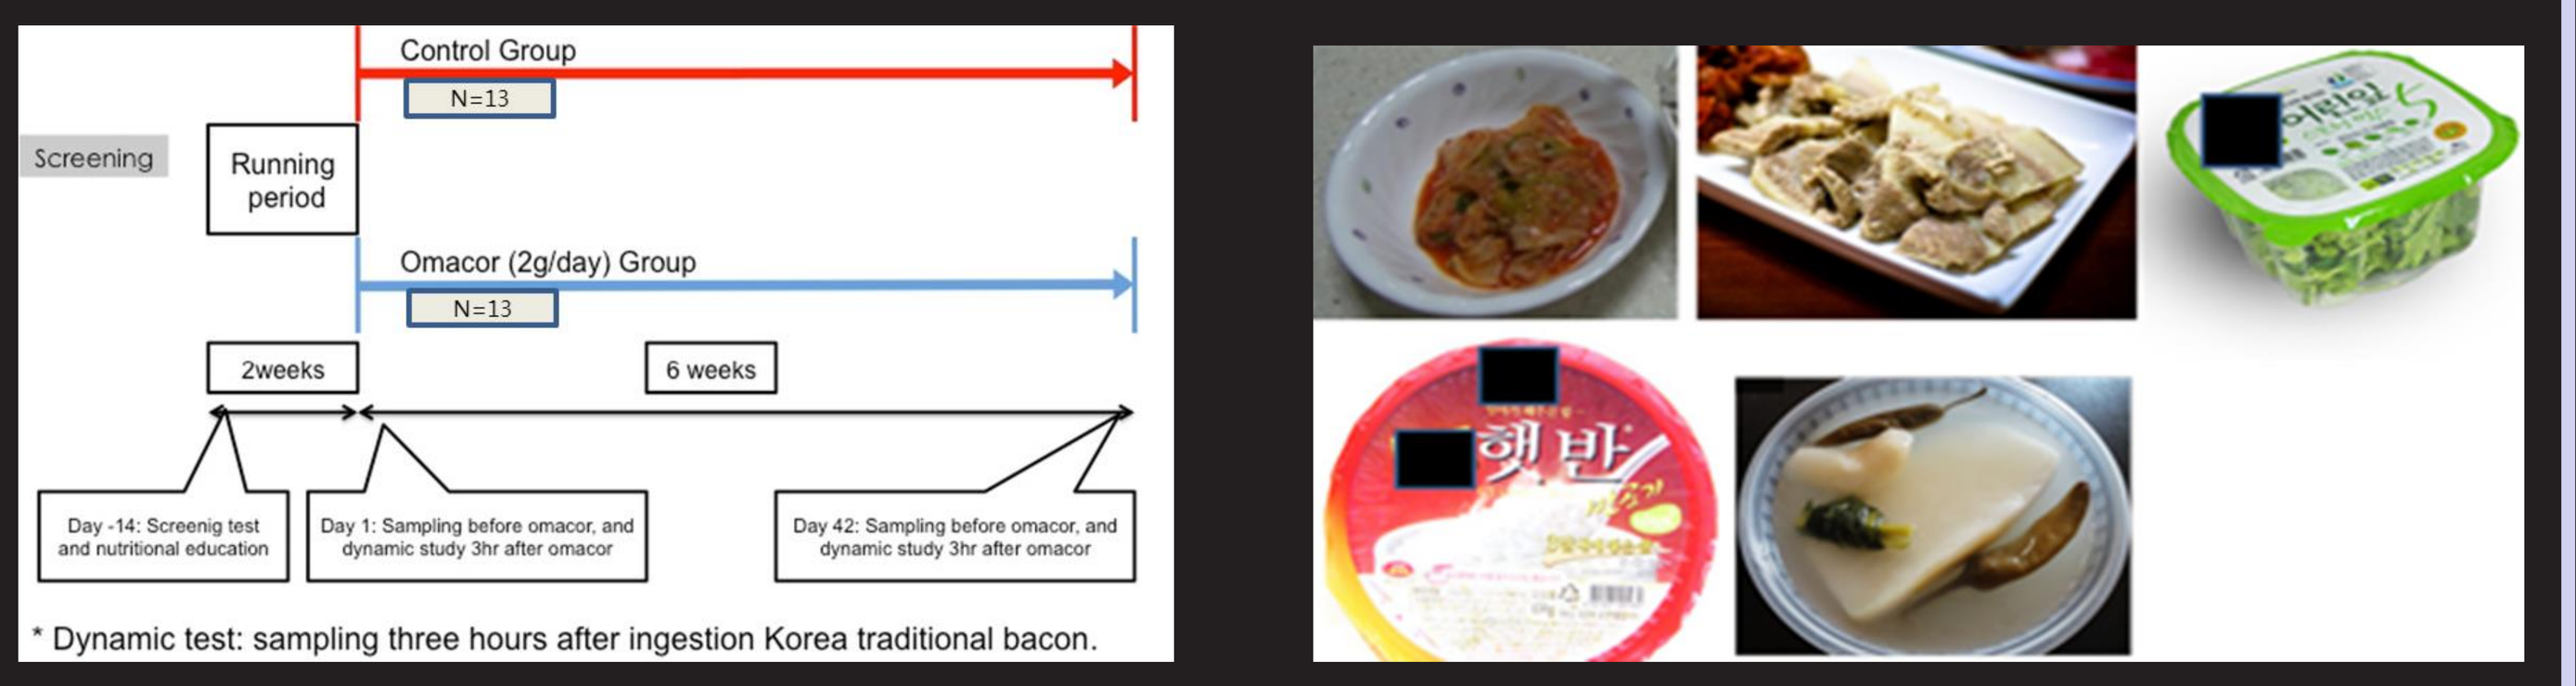
Control Group
N=13
Screening
Running period
Omacor (2g/day) Group
N=13
2weeks
6 weeks
Day -14: Screenig test and nutritional education
Day 1: Sampling before omacor, and dynamic study 3hr after omacor
Day 42: Sampling before omacor, and dynamic study 3hr after omacor
* Dynamic test: sampling three hours after ingestion Korea traditional bacon.

Figure 1. Right: Study protocol. Left: standard test meal with 710 kcal

Subjects were asked to complete a food frequency questionnaires and a 24 h dietary recall interview was conducted on morning of the study. The standardized test meal included 710 kcal with steamed folk.

All patients were educated for importance of lifestyle modification after getting informed consent. Subjects attended the study centrer after an overnight fast and were asked to refrain from alcohol consumption during 48hr before visit for the study.

All data were presented as mean ± SD. P values were obtained by using Mann-whitney test.

Results

	Patients with omega-3	Patients without omega-3
Number	13	13
Gender, Female	5 (39)	6 (50)
Age (y)	49.9 ± 10.4	52.2 ± 12.1
Height (cm)	163.5 ± 6.9	162.5 ± 8.4
Weight (kg)	72.0 ± 11.5	69.7 ± 11.1
Waist circumference (cm)	93.0 ± 5.4	95.2 ± 10.4
SBP (mmHg)	132.8 ± 16.3	129.8 ± 7.3
DBP (mmHg)	83.9 ± 8.0	81.8 ± 11.8
Presence of Diabetes	5 (38)	4 (31)
Presence of Hypertension	4 (31)	6(46)
anti-dyslipidemic medication	3 (23)	3 (23)
FBG (mg/dL)	105.5 ± 25.7	108.2 ± 27.9

Table 1. Baseline characteristics. Data are presented as number(%) or mean ± SD.

	Patients with omega-3			Patients without omega-3		
Fasting	**Day 1**	**Day 42**	**P value**	**Day 1**	**Day 42**	**P value**
TC (mg/dL),	171.5 ± 43.0	175.3 ± 50.2	0.65	214.0 ± 37.0	211.1 ± 48.3	0.79
TG (mg/dL)	258.2 ± 117.4	254.3 ± 246.4	0.046	271.2 ± 221.7	255.1 ± 198.0	0.79
HDL (mg/dL)	41.6 ± 10.3	44.1 ± 11.4	0.124	44.2 ± 13.0	51.0 ± 17.0	0.075
LDL (mg/dL)	100.4 ± 46.0	97.2 ± 51.8	0.753	140.5 ± 39.8	134.2 ± 49.6	0.155
Postprandial	**Day 1**	**Day 42**	**P value**	**Day 1**	**Day 42**	**P value**
TC (mg/dL),	173.3 ± 41.7	172.0 ± 48.4	0.807	214.6 ± 38.6	206.7 ± 48.4	0.230
TG (mg/dL)	357.3 ± 120.9	277.2 ± 157.6	0.033	343.7 ± 195.1	324.7 ± 226.5	0.859
HDL (mg/dL)	40.7 ± 9.5	42.5 ± 11.4	0.311	43.4 ± 13.4	44.9 ± 13.5	0.689
LDL (mg/dL)	96.8 ± 44.9	94.5 ± 50.6	0.807	137.7 ± 37.3	127.8 ± 48.2	0.142

Table 2. Fasting and 3-hr postprandial lipid parameters. P values were calculated by comparing data between Day 1 and Day 42, intragroup analysis. TC: total cholesterol, TG: triglyceride, HDL: high density lipoprotein cholesterol, LDL: low density lipoprotein cholesterol

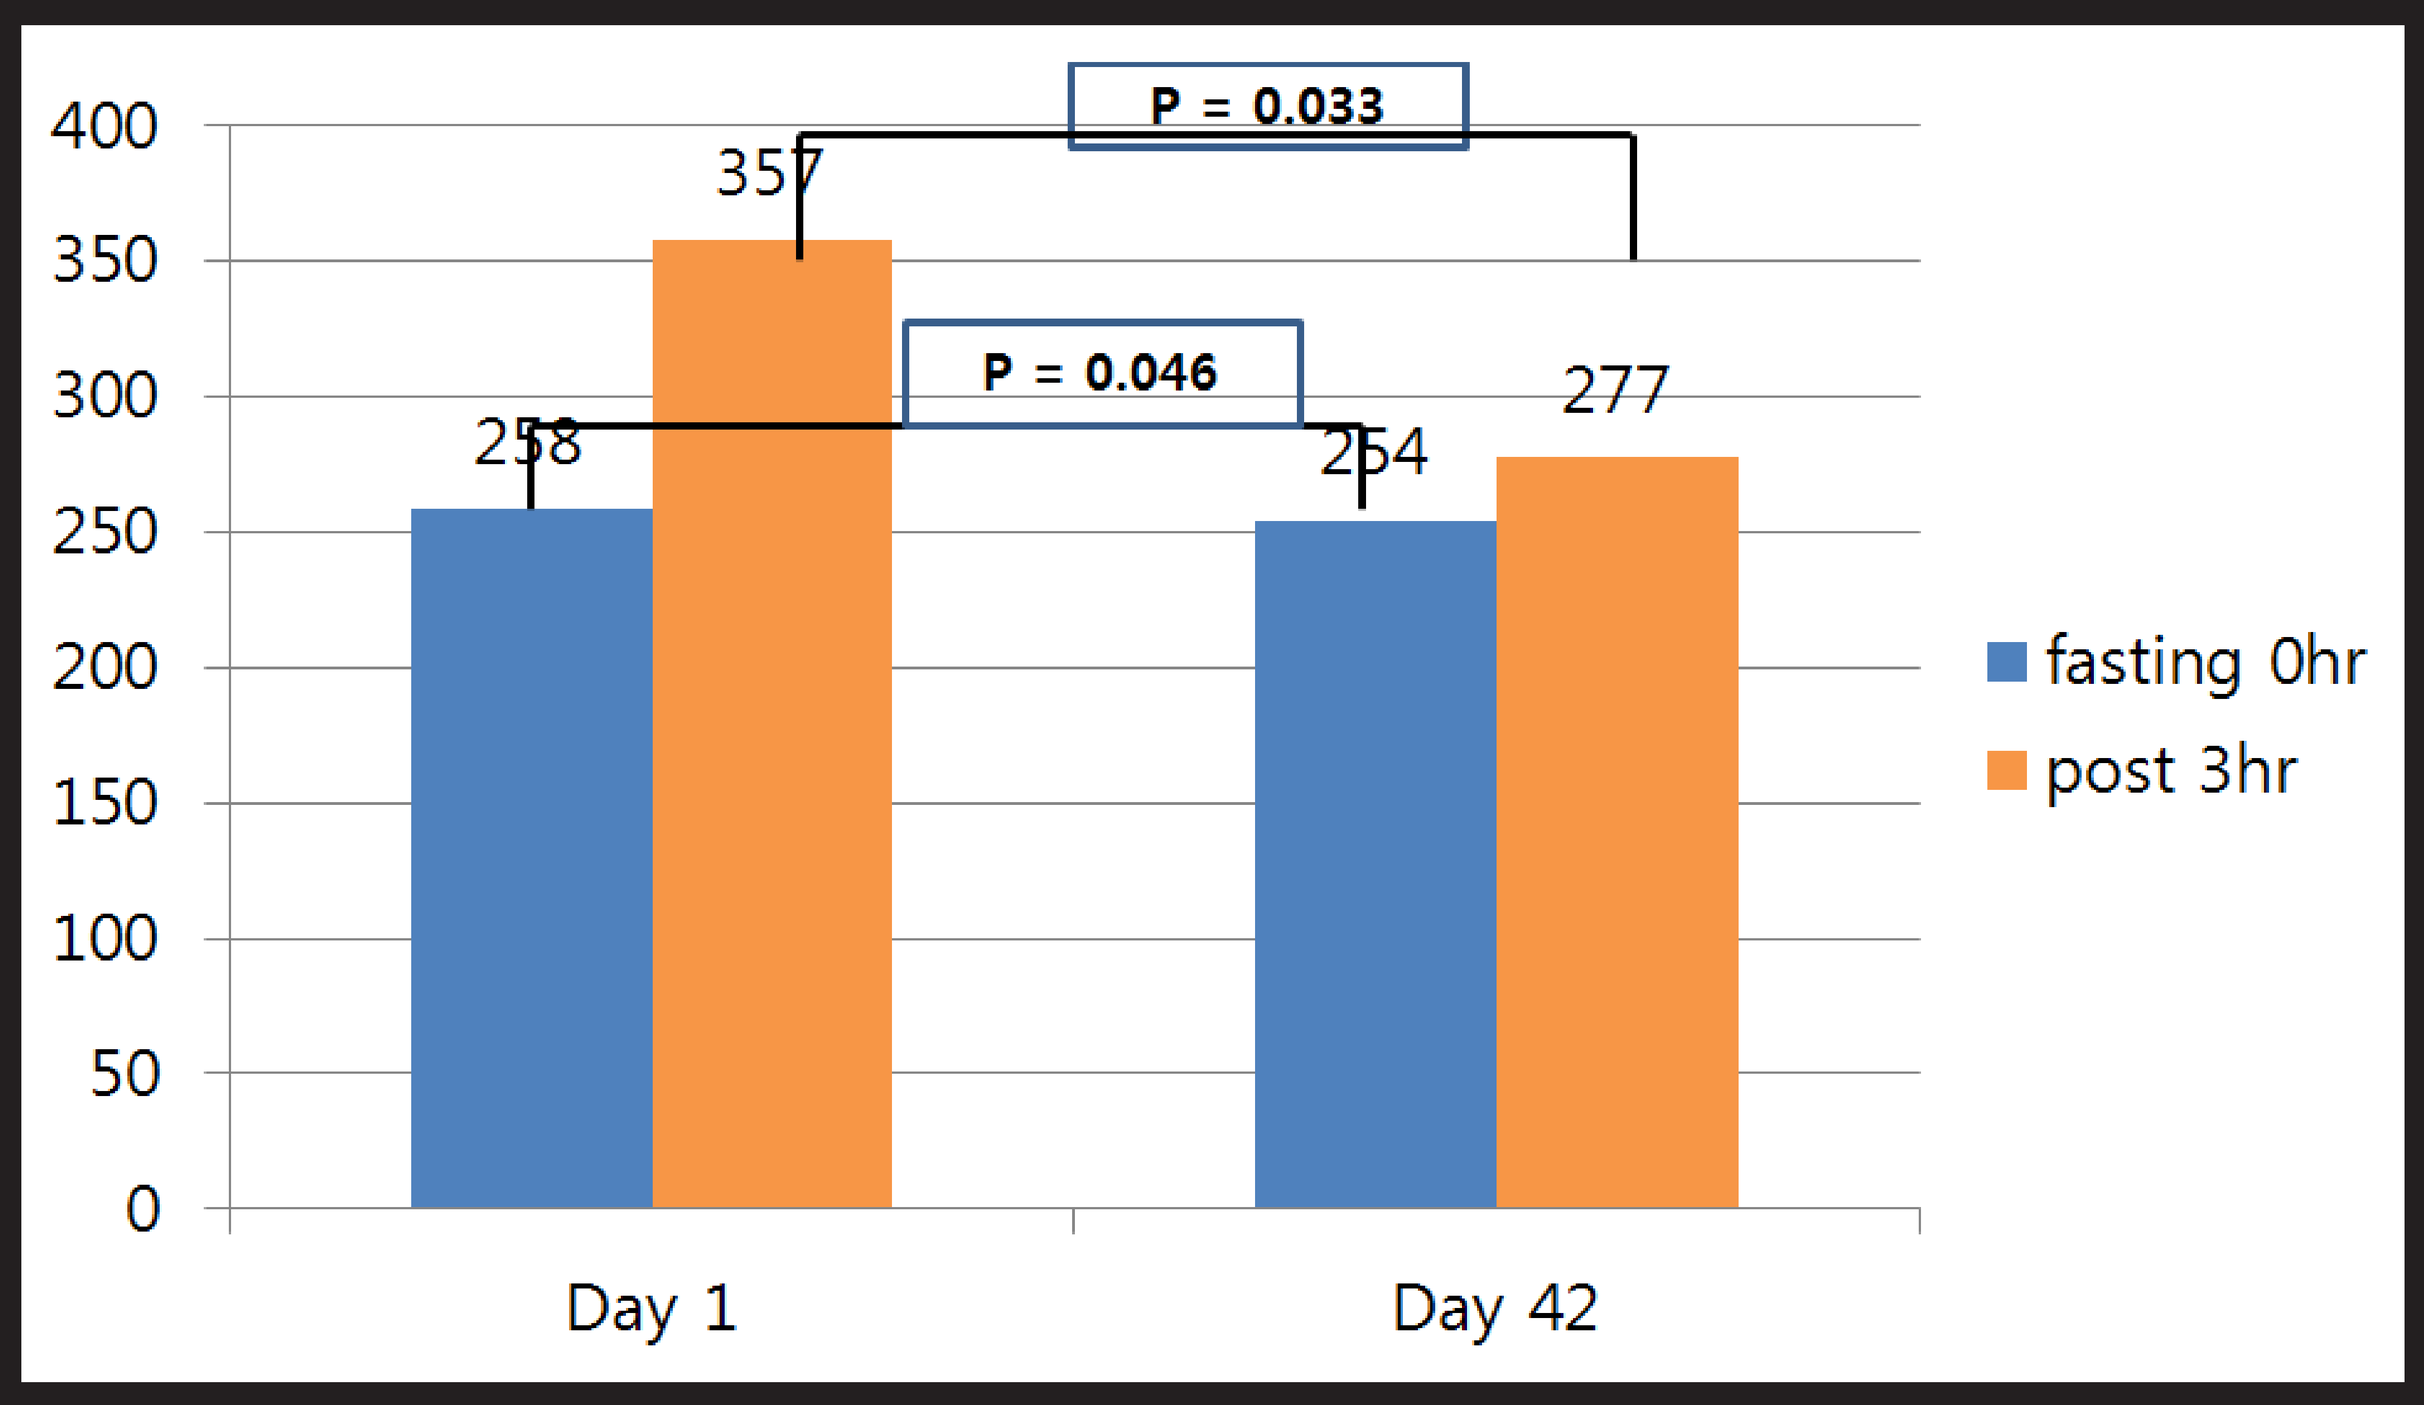
400
350
300
250
200
150
100
50
0
P = 0.033
P = 0.046
357
258
254
277
Day 1
Day 42
fasting 0hr
post 3hr

Figure 2.TG variation in the group with omega- 3 medication during dyanmic test.

In intragroup analysis, treatment group with omega-3 fatty acids showed significant decrease on fasting TG (258.2 mg/dl vs. 254.2 mg/dl, P = 0.046) and especially post-prandial TG (357.3 mg/dl vs. 277.2 mg/dl, P = 0.033), while control group showed no significant difference during follow-up (271.2 mg/dl vs. 255.1 mg/dl, P = 0.790 for fasting TG and 343.7 mg/dl vs. 324.7 mg/dl for post-prandial TG). However, in intergroup analysis, there was no significant difference between two groups in fasting and post-prandial TG (% change: P = 0.287 and 0.303, respectively). There was no significant improvement in other lipid profiles. No significant adverse events were registered during this study.

Conclusion

The omega-3 fatty acids achieved a significant reduction of fasting and post-prandial triglycerides without adverse reactions. Because the trend of improvement in post-prandial TG appeared in the treatment group than control group, further study including more subjects will be needed.

(This study was supported by Gun-il Pharmacy, Seoul, Korea)

References

1. Rapp JH, et al. Arterioscler Thromb. 1994;14(11):1767-1774.
2. Proctor SD, et al. Eur J Clin Invest. 1998;28(6):497-503
3. Bansal S, et al. JAMA. 2007;298:309-316
4. Nordestgaard BG al. JAMA 2007;298:299-308

17 ECE 614-EP
Obesity and cardiovascular endocrinology
Won Jun Kim
DOI: 10.3252/pso.eu.17ece.2015
Poster presented at: ECE2015 16-20 May 2015 DUBLIN, IRELAND 17th EUROPEAN CONGRESS OF ENDOCRINOLOGY
PosterSessionOnline.com