Growth hormone therapy and effect on ovarian function and morphology in short prepubertal SGA girls

Jeanette Tinggaard[1], Rikke Beck Jensen[1], Karin Sundberg[2], Katharina M. Main[1], Niels Birkebaek[3], Peter Christiansen[1], Annie Ellermann[4], Kirsten Holm[5], Eva Mosfeldt-Jeppesen[6], Nina Saurbrey[6], Britta Kremke[7], Pawel Marcinski[8], Rune Weis Naeraa[3], Carsten Pedersen[9], Brigitte Scherling[10], Ebbe Thisted[11], the NESGAS group*, Anders Juul[1]

1 University Department of Growth and Reproduction, Rigshospitalet, Copenhagen, Denmark, **2** Department of Fetal Medicine, Rigshospitalet, Copenhagen, Denmark**, 3** Department of Pediatrics, Aarhus University Hospital Skejby, Denmark, **4** Department of Pediatrics, Holbæk sygehus, Denmark, **5** Department of Pediatrics, Hillerød sygehus, Denmark**, 6** Department of Pediatrics, Glostrup Hospital, Denmark**, 7** Department of Pediatrics, Randers sygehus, Denmark**, 8** Department of Pediatrics, Vendsyssel sygehus Hjørring, Denmark, **9** Department of Pediatrics, Kolding sygehus, Denmark, **10** Department of Pediatrics, Roskilde sygehus, Denmark, **11** Department of Pediatrics, Sønderborg Sygehus, Denmark.

Disclosure statement: This study was supported by Novo Nordisk A/S

Background

Girls born small for gestational age (SGA) are at risk of early pubertal maturation and development of PCOS. Growth Hormone (GH) receptors are present in ovaries, and GH may have a physiological role for ovarian function, but little is known about the effect of GH therapy on ovarian function in short girls born SGA. The objective of this study was to evaluate ovarian morphology and function during GH therapy.

Methods

Clinical characteristics, reproductive hormones and ultrasonographic examination of the internal genitalia were determined in 18 prepubertal girls during the first three years of GH therapy in a Danish sub-study of the North European SGA study (NESGAS); a multinational, randomised, longitudinal study of GH therapy in short prepubertal children born SGA.

Results

Median age at baseline was 4.9 years (4.5-7.2) (Table 1). Bone age advanced significantly during three years of treatment (P=0.007), but did not exceed chronological age. Uterine and ovarian volume increased significantly (P=0.033 and P=0.005, respectively), but remained low within the normal reference ranges (Figure 1, left panel). Ovarian follicles became visible in 69% of girls compared to 28% of girls before GH therapy. No significant changes in follicular number or size were found. Precocious puberty was observed in one girl and another girl showed signs of a multicystic ovary verified by ultrasound. Anti-Müllerian Hormone tended to cluster in the lower part of the reference range, although a significant increase during three years of GH-treatment was observed (P=0.028) (figure 1, right panel). SHBG decreased during the first year of GH therapy (P<0.001) and remained low, while an increase in Δ4-androstenedione and DHEAS was found. Inhibin B increased significantly, but no significant changes in FSH, LH, estradiol or inhibin A were found.

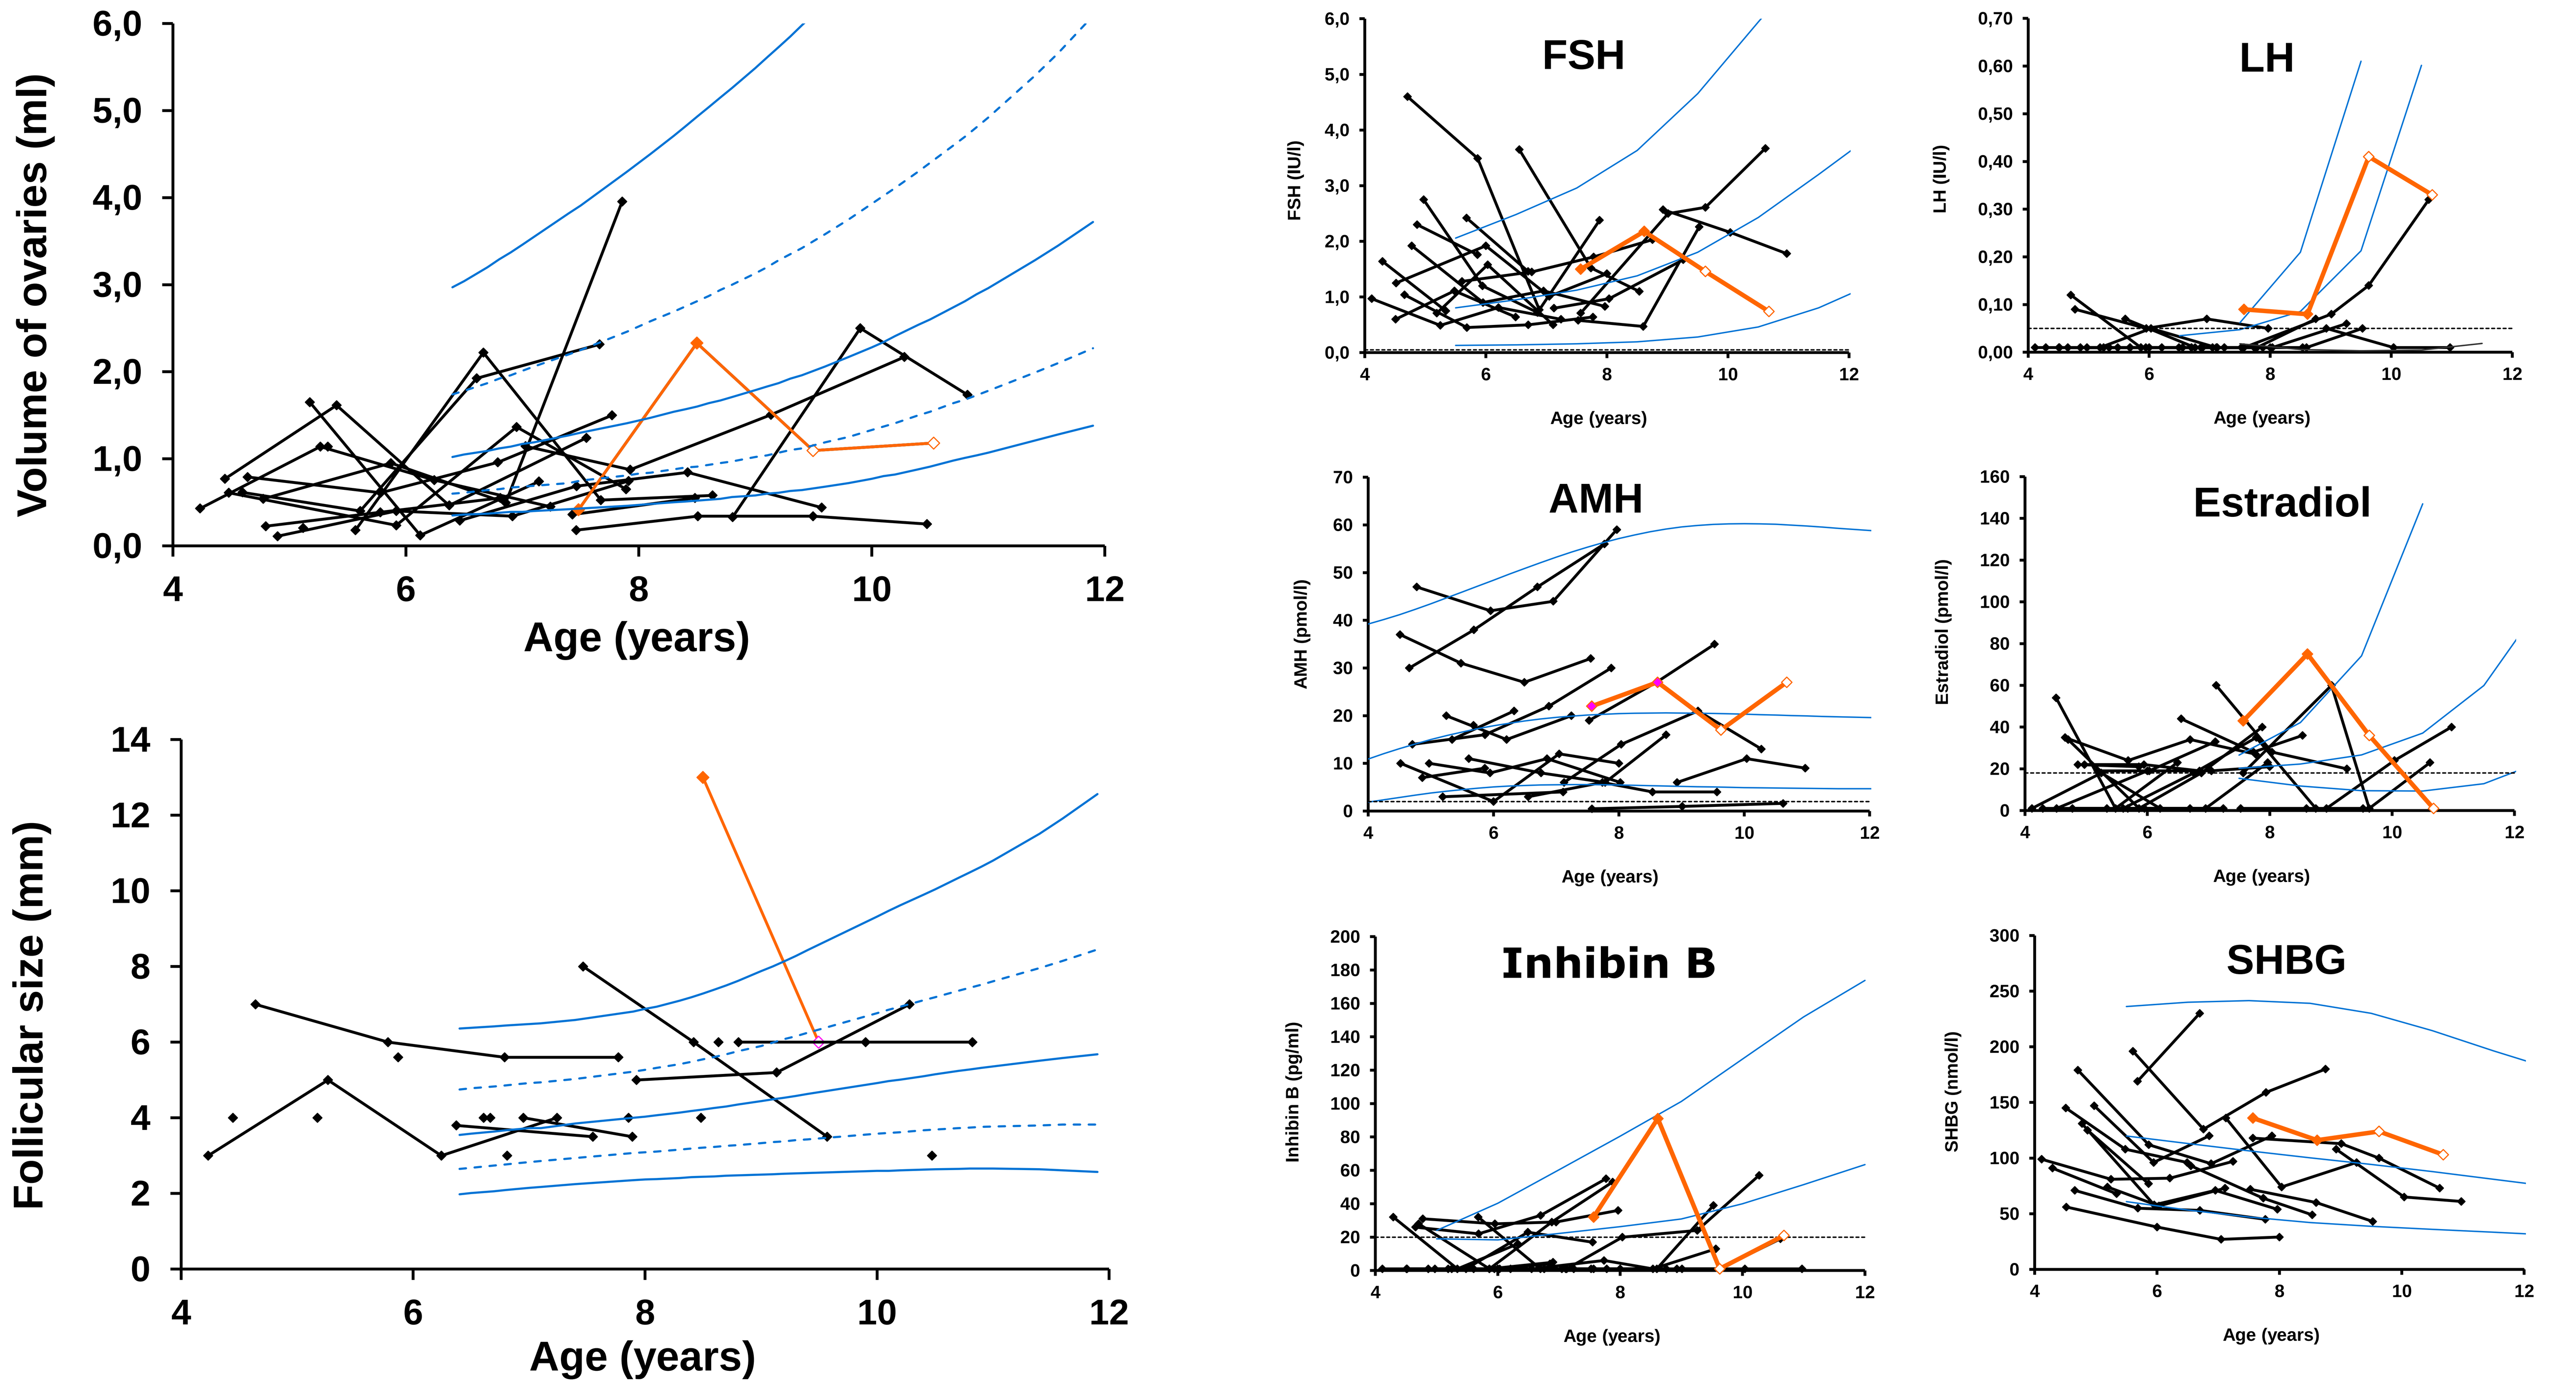
Volume of ovaries (ml)
Age (years)
Follicular size (mm)
Age (years)
FSH
FSH (IU/l)
Age (years)
LH
LH (IU/l)
Age (years)
AMH
AMH (pmol/l)
Age (years)
Estradiol
Estradiol (pmol/l)
Age (years)
Inhibin B
Inhibin B (pg/ml)
Age (years)
SHBG
SHBG (nmol/l)
Age (years)

Figure 1, left panel; Volume of ovaries (upper figure) and follicular size (lower figure) according to age during GH therapy in 18 short prepubertal SGA girls. **Figure 1, right panel:** Circulating serum levels of FSH, LH, AMH, Estradiol, Inhibin B and SHBG according to age during GH therapy in 18 short prepubertal SGA girls. Solid blue lines represent mean +/- 2 SD and dotted blue lines represent +/- 1 SD. One girl who presented with precocious puberty is highlighted in colour with filled squares before and blank squares after treatment with a GnRH agonist.

	Baseline (n=18)	1 year (n=17)	2 year (n=17)	3 year (n= 14)
Chronological Age (yr)	4.91 (4.51-7.22)	5.95 (5.58-8.32)	6.95 (6.59-9.39)	7.98 (7.50-9.74)
Bone age (BoneXpert)	3.27 (2.09-5.74)	4.49 (3.23-6.81)	6.56 (5.41-7.87)	7.20 (6.22-8.71)
BA-CA	-1.64 (-2.53 to -1.13)	-1.59 (-2.17 to -1.22)	-1.05 (-1.78 to -0.23)*	-1.14 (-1.71 to 0.42)#
Weight (SDS)	-2.88 (-3.24 to -1.67)	-1.72 (-2.50 to -0.56)*	-1.38 (-2.30 to 0.15)*	-1.57 (-2.08 to 0.10)*#
Height (SDS)	-2.95 (-3.47 to -2.62)	-1.97 (-2.69 to -1.47)*	-1.56 (-2.39 to -0.98)*	-1.41 (-2.00 to 0.35)*#
BMI (SDS)	-0.60 (-1.71 to -0.25)	-0.71 (-1.83 to -0.61)	-0.43 (-1.81 to 0.69)	-0.79 (-1.93 to -0.64)
Tanner stage I/II	18/0	16/1	15/2	13/1
Uterine volume (ml)	1.05 (0.68-1.56)	1.22 (0.88-2.32)*	1.50 (1.04-2.20)	1.72 (0.88-2.45) #
Ovarian volume (ml)	0.43 (0.31-0.69)	0.69 (0.39-1.63)	0.76 (0.47-1.30)	0.90 (0.62-1,84) #
Largest follicle (mm)	5.0 (3.3-6.8)	5.3 (4.0-6.5)	4.6 (3.6-6.0)	4.0 (3.5-5.9)
FSH (IU/l)	1.50 (0.89-2.50)	1.33 (0.73-2.04)	1.11 (0.75-1.70)	0.98 (0.63-1.64)
LH (IU/l)	0.01 (0.01-0.06)	0.01 (0.01-0.01)	0.01 (0.01-0.05)*	0,05 (0.01-0.06)
AMH (pmol/l)	11 (6-22)	14 (8-27)	21 (11-35)	20 (8-31) #
Estradiol (pmol/l)	18 (1-39)	19 (1-26)	20 (10-36)	20 (14-26)
SHBG (nmol/l)	125 (92-146)	74 (62-110)*	84 (55-98)	80 (52-99) #
DHEAS (mmol/l)	0.47 (0.01-0.56)	0.51 (0.01-1.23)*	0.79 (0.71-2.05)*	1.36 (1.26-1.61)* #
Androstenedione (mmol/l)	0.01 (0.01-0.01)	0.01 (0.01-1.87)*	0.01 (0.01-1.50)	1.19 (0.01-1.93) #
Testosterone (nmol/l)	0.01 (0.01-0.01)	0.01 (0.01-0.01)	0.01 (0.01-0.01)	0.01 (0.01-0.01)

Table 1 Clinical characteristics and hormone analyses at baseline and after one, two and three years of GH treatment. Results are presented as medians (interquartile range). *P<0.05 compared to previous year by Wilcoxon sign rank test, #P<0.05 for values at three years compared to baseline.

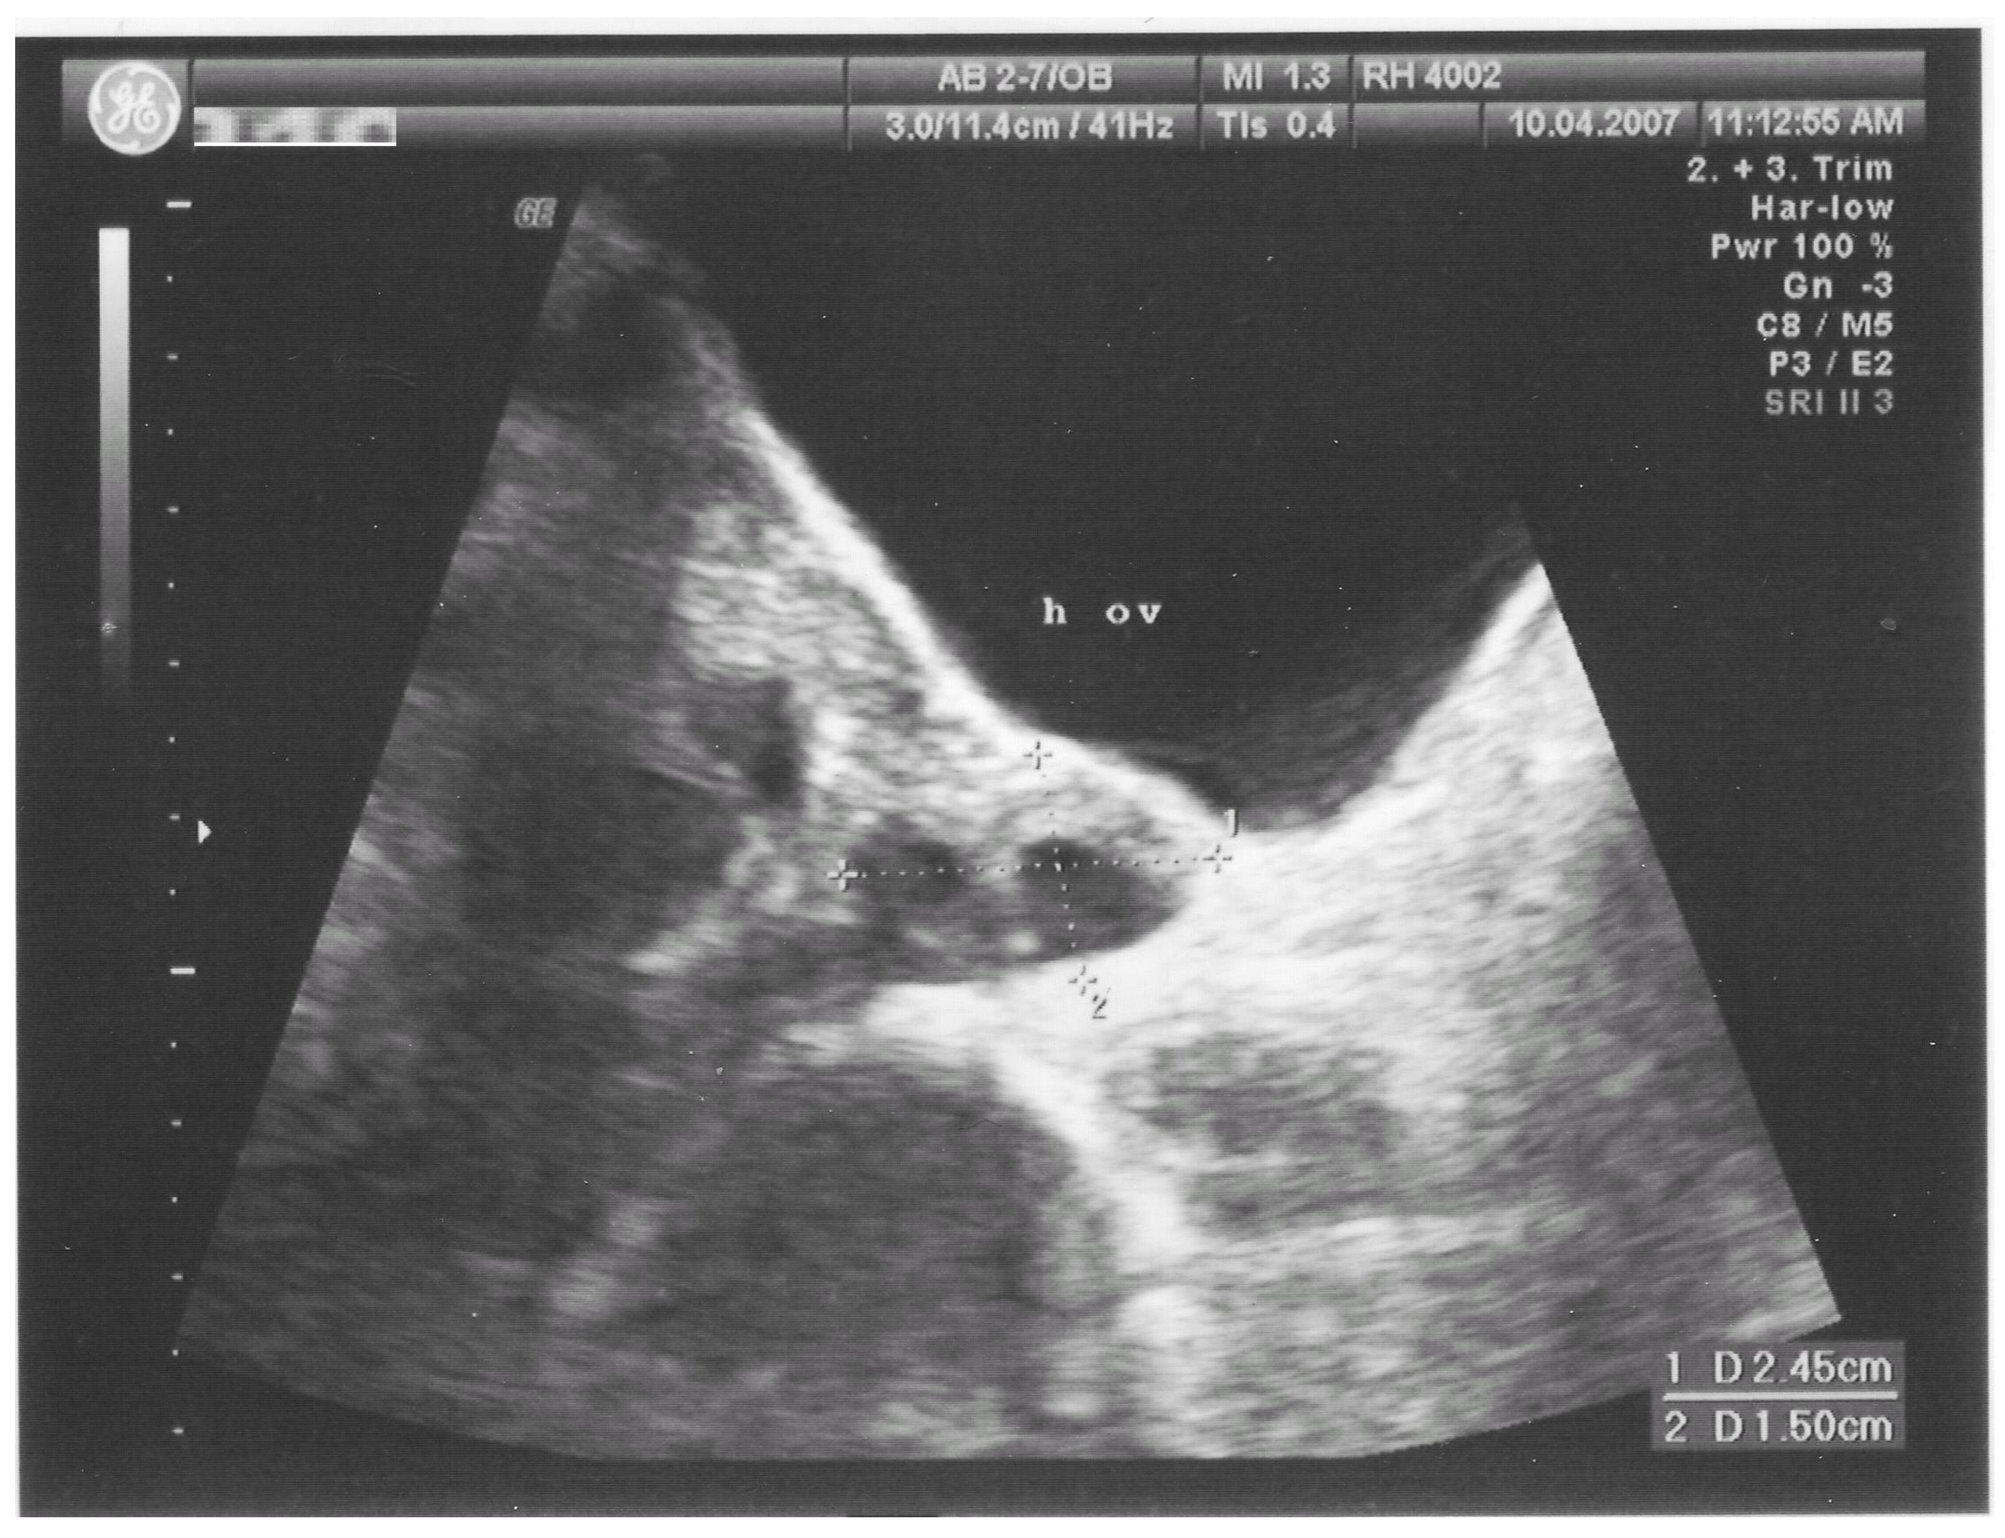
AB 2-7/OB
MI 1.3
RH 4002
3.0/11.4cm / 41Hz
TIs 0.4
10.04.2007
11:12:55 AM
2. + 3. Trim
Har-low
Pwr 100 %
Gn -3
C8 / M5
P3 / E2
SRI II 3
h ov
1 D 2.45cm
2 D 1.50cm

Figure 2. Ultrasonographic examination of the right ovary showing follicular activity

Conclusions

GH therapy was associated with marked increase in height, and considered safe. Size and morphology of internal genitals increased proportionally to the increased body size during GH therapy. However, 2 of 18 girls developed precocious puberty (central) or multicystic ovary, respectively. It remains uncertain whether or not these findings were associated with the concomitant GH therapy, or independent phenomena.

*) The NESGAS group include the following researchers (listed according to center and in alphabetical order): **Birmingham**: Mandy Aspinall, Jeremy Kirk, Tracey Kirkwood **Cambridge**: David Dunger*, Catherine Fullah **Dublin**: Hillary Hoey*, Susan O'Connell, Elaine O'Mullane, Edna Roche **Glasgow**: Malcolm Donaldson*, Sheena McGowern **Malmoe**: Sten Ivarsson*, Helena Larsson, Carina Persson **Lund**: Maria Elfving, Lena Rollof **Stockholm**: Lo Neumeyer, Olle Söder*

THE UNIVERSITY OF DUBLIN

UNIVERSITY OF CAMBRIDGE

UNIVERSITY OF COPENHAGEN

UNIVERSITY of GLASGOW

UNIVERSITY OF BIRMINGHAM

Karolinska Institutet

LUND UNIVERSITY